SIG-1191™: A novel hydrating cosmetic functional ingredient (CFI) for suborbital hyperpigmentation (under eye dark circles)

José R. Fernández[1], Karl Rouzard[1], Michael Voronkov[1], Jason Healy[1], Kristen L. Huber[1], Corey Webb[1], Jeffry B. Stock[1,2]
Maxwell Stock[1], Joel S. Gordon[1], Eduardo Pérez[1]
[1]Signum Dermalogix, 133 Wall Street, Princeton, NJ; [2]Princeton University, Department of Molecular Biology, Princeton, NJ

Abstract

The appearance of dark circles around the eyes is due in part to the lack of hydration and accumulation of heme, which is pro-oxidative and pro-inflammatory. Isoprenylcysteine (IPC) compounds have been shown to regulate the responses of inflammatory cells and increase expression and production of Aquaporin-3 (AQP3), which promotes skin hydration. We report here an IPC derivative and cosmetic functional ingredient (CFI), SIG-1191™, possesses antioxidant activity in dermal fibroblasts. Similar to previous IPC CFIs, Arazine® and SIG-1273™, SIG-1191™ also demonstrates anti-inflammatory properties *in vitro* by reducing UVB-induced pro-inflammatory cytokine production (IL-6 and TNF-α) in primary human keratinocytes. In addition to its anti-inflammatory and antioxidant properties, SIG-1191™ potentially targets heme metabolism by modulating Heme oxygenase (HO-1) expression. HO-1 regulates heme metabolism from leaky blood vessels of the suborbital dermis. Accumulated heme generates the appearance of violet pigment in this area, producing dark coloration though the skin and the production of free radicals and inflammation. SIG-1191™ dose-dependently stimulates HO-1 gene and protein expression in both human dermal microvascular endothelial cells (HDMECs) as well as when applied topically in a three dimensional (3D) reconstituted human skin equivalent. Additionally, SIG-1191™ dose-dependently increased HO-1 protein levels, as determined by specific nuclear staining, in both keratinocytes and fibroblasts of 3D skin equivalents Altogether, these studies demonstrate SIG-1191™ is a novel CFI that potentially increases microvascular heme metabolism by increasing HO-1 and possesses antioxidant, anti-inflammatory and hydrating properties to help reduce the appearance of dark circles under the eye.

Fig 1. Suborbital hyperpigmentation (aka dark circles)

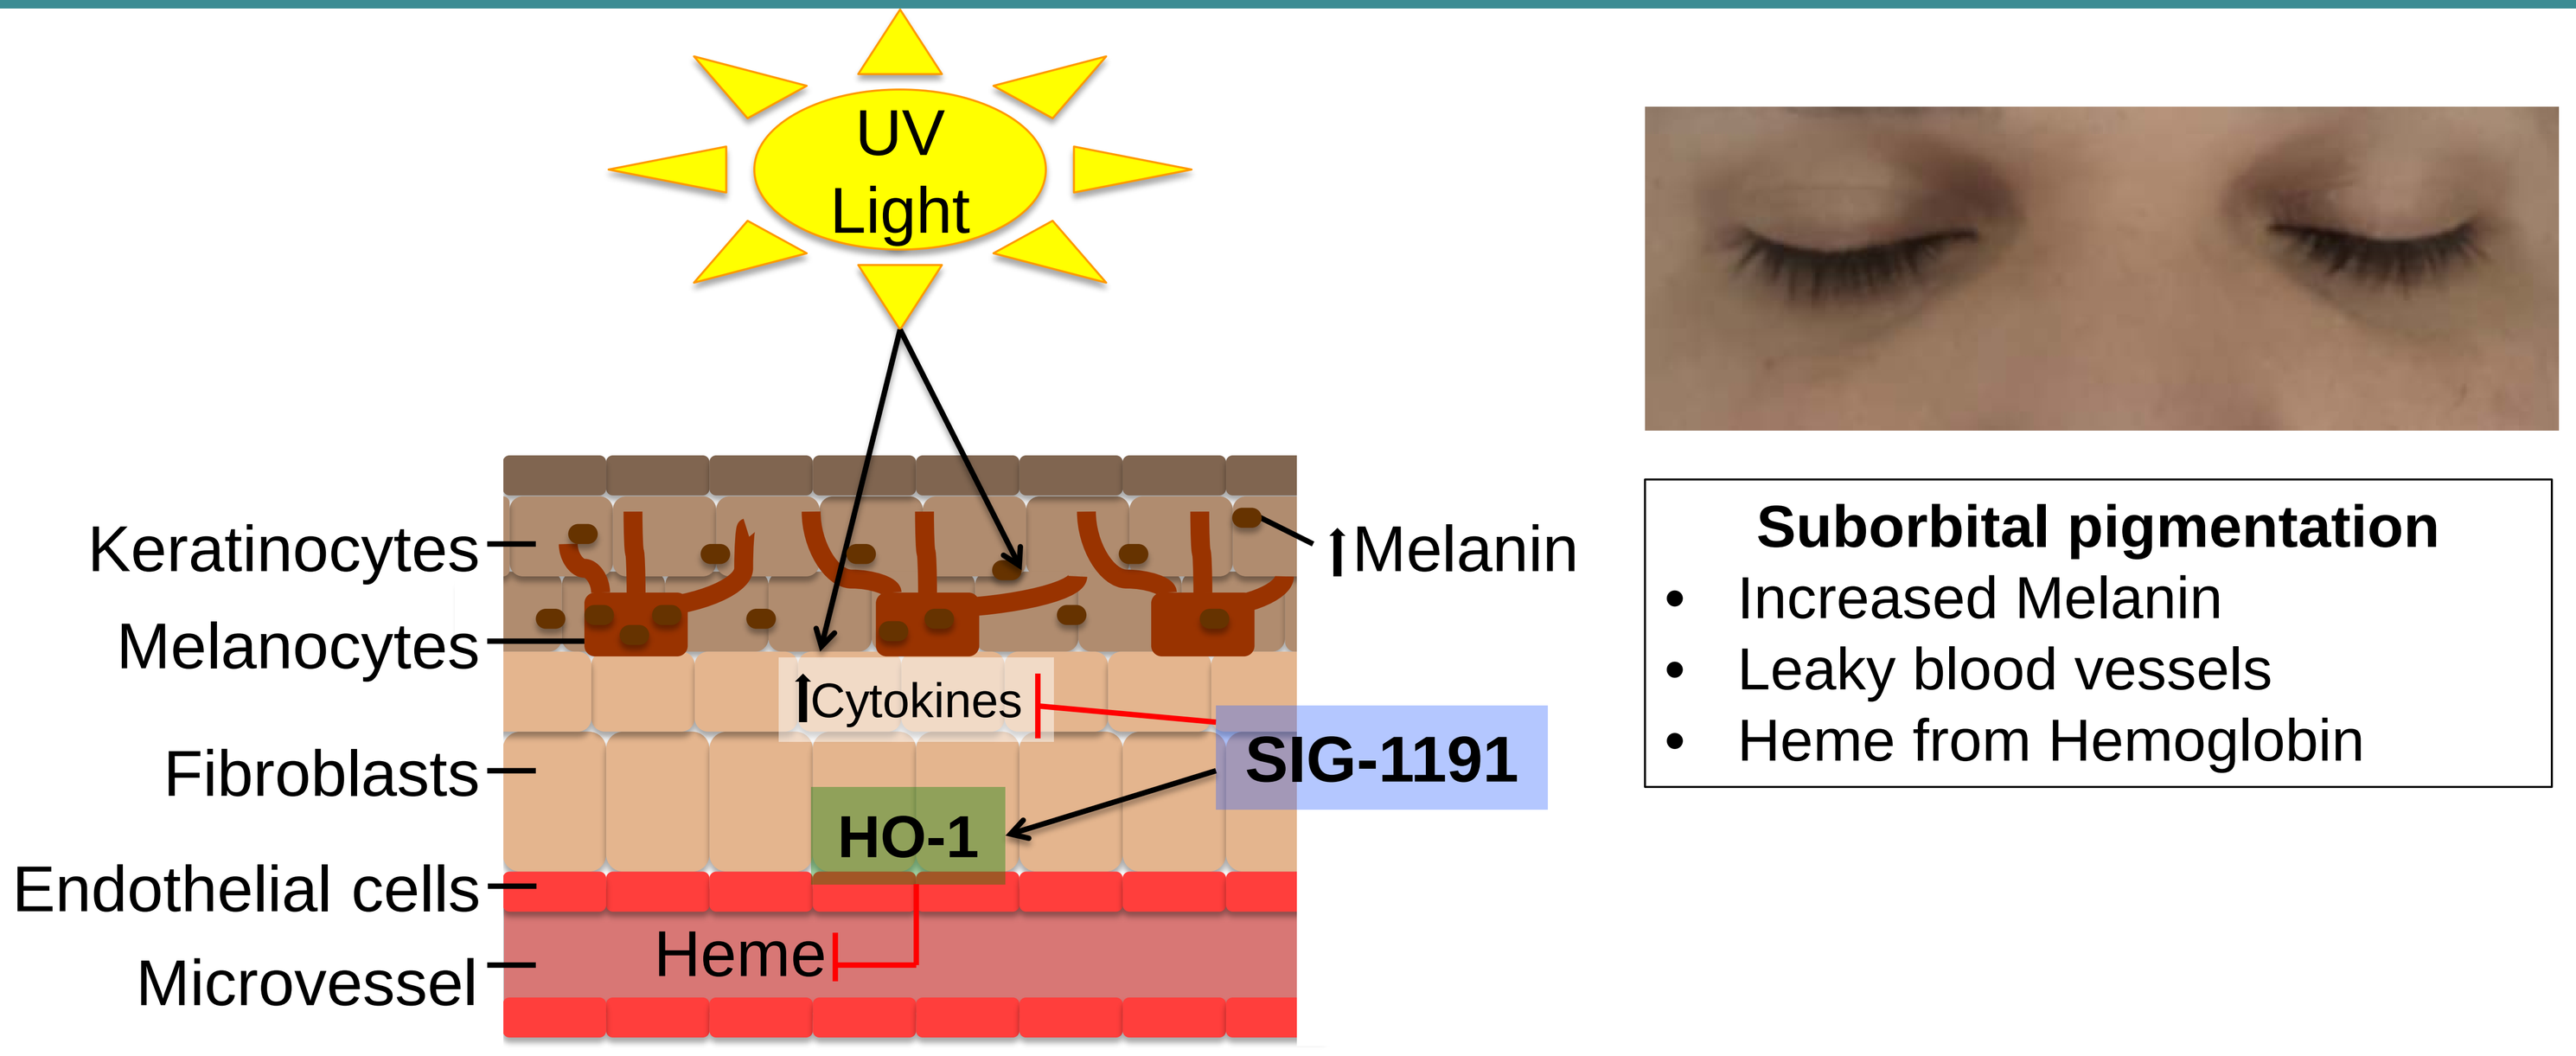
UV Light
Keratinocytes
Melanocytes
Fibroblasts
Endothelial cells
Microvessel
Melanin
Cytokines
SIG-1191
HO-1
Heme
Suborbital pigmentation
• Increased Melanin
• Leaky blood vessels
• Heme from Hemoglobin

Excessive pigmentation in the suborbital skin is caused by post-inflammatory hyperpigmentation, heme accumulation and/or triggered by environmental exposure to UV light, leading to release pro-inflammatory cytokines and absorption of melanin pigment. Heme pigment produced after blood leakage is observed and is regulated by Heme oxygenase (HO-1). SIG-1191™ is a novel, multi-functional CFI that inhibits pro-inflammatory cytokine production and increases HO-1 expression for heme metabolism.

Fig 2. SIG-1191™ increases keratinocyte AQP3 gene and protein expression

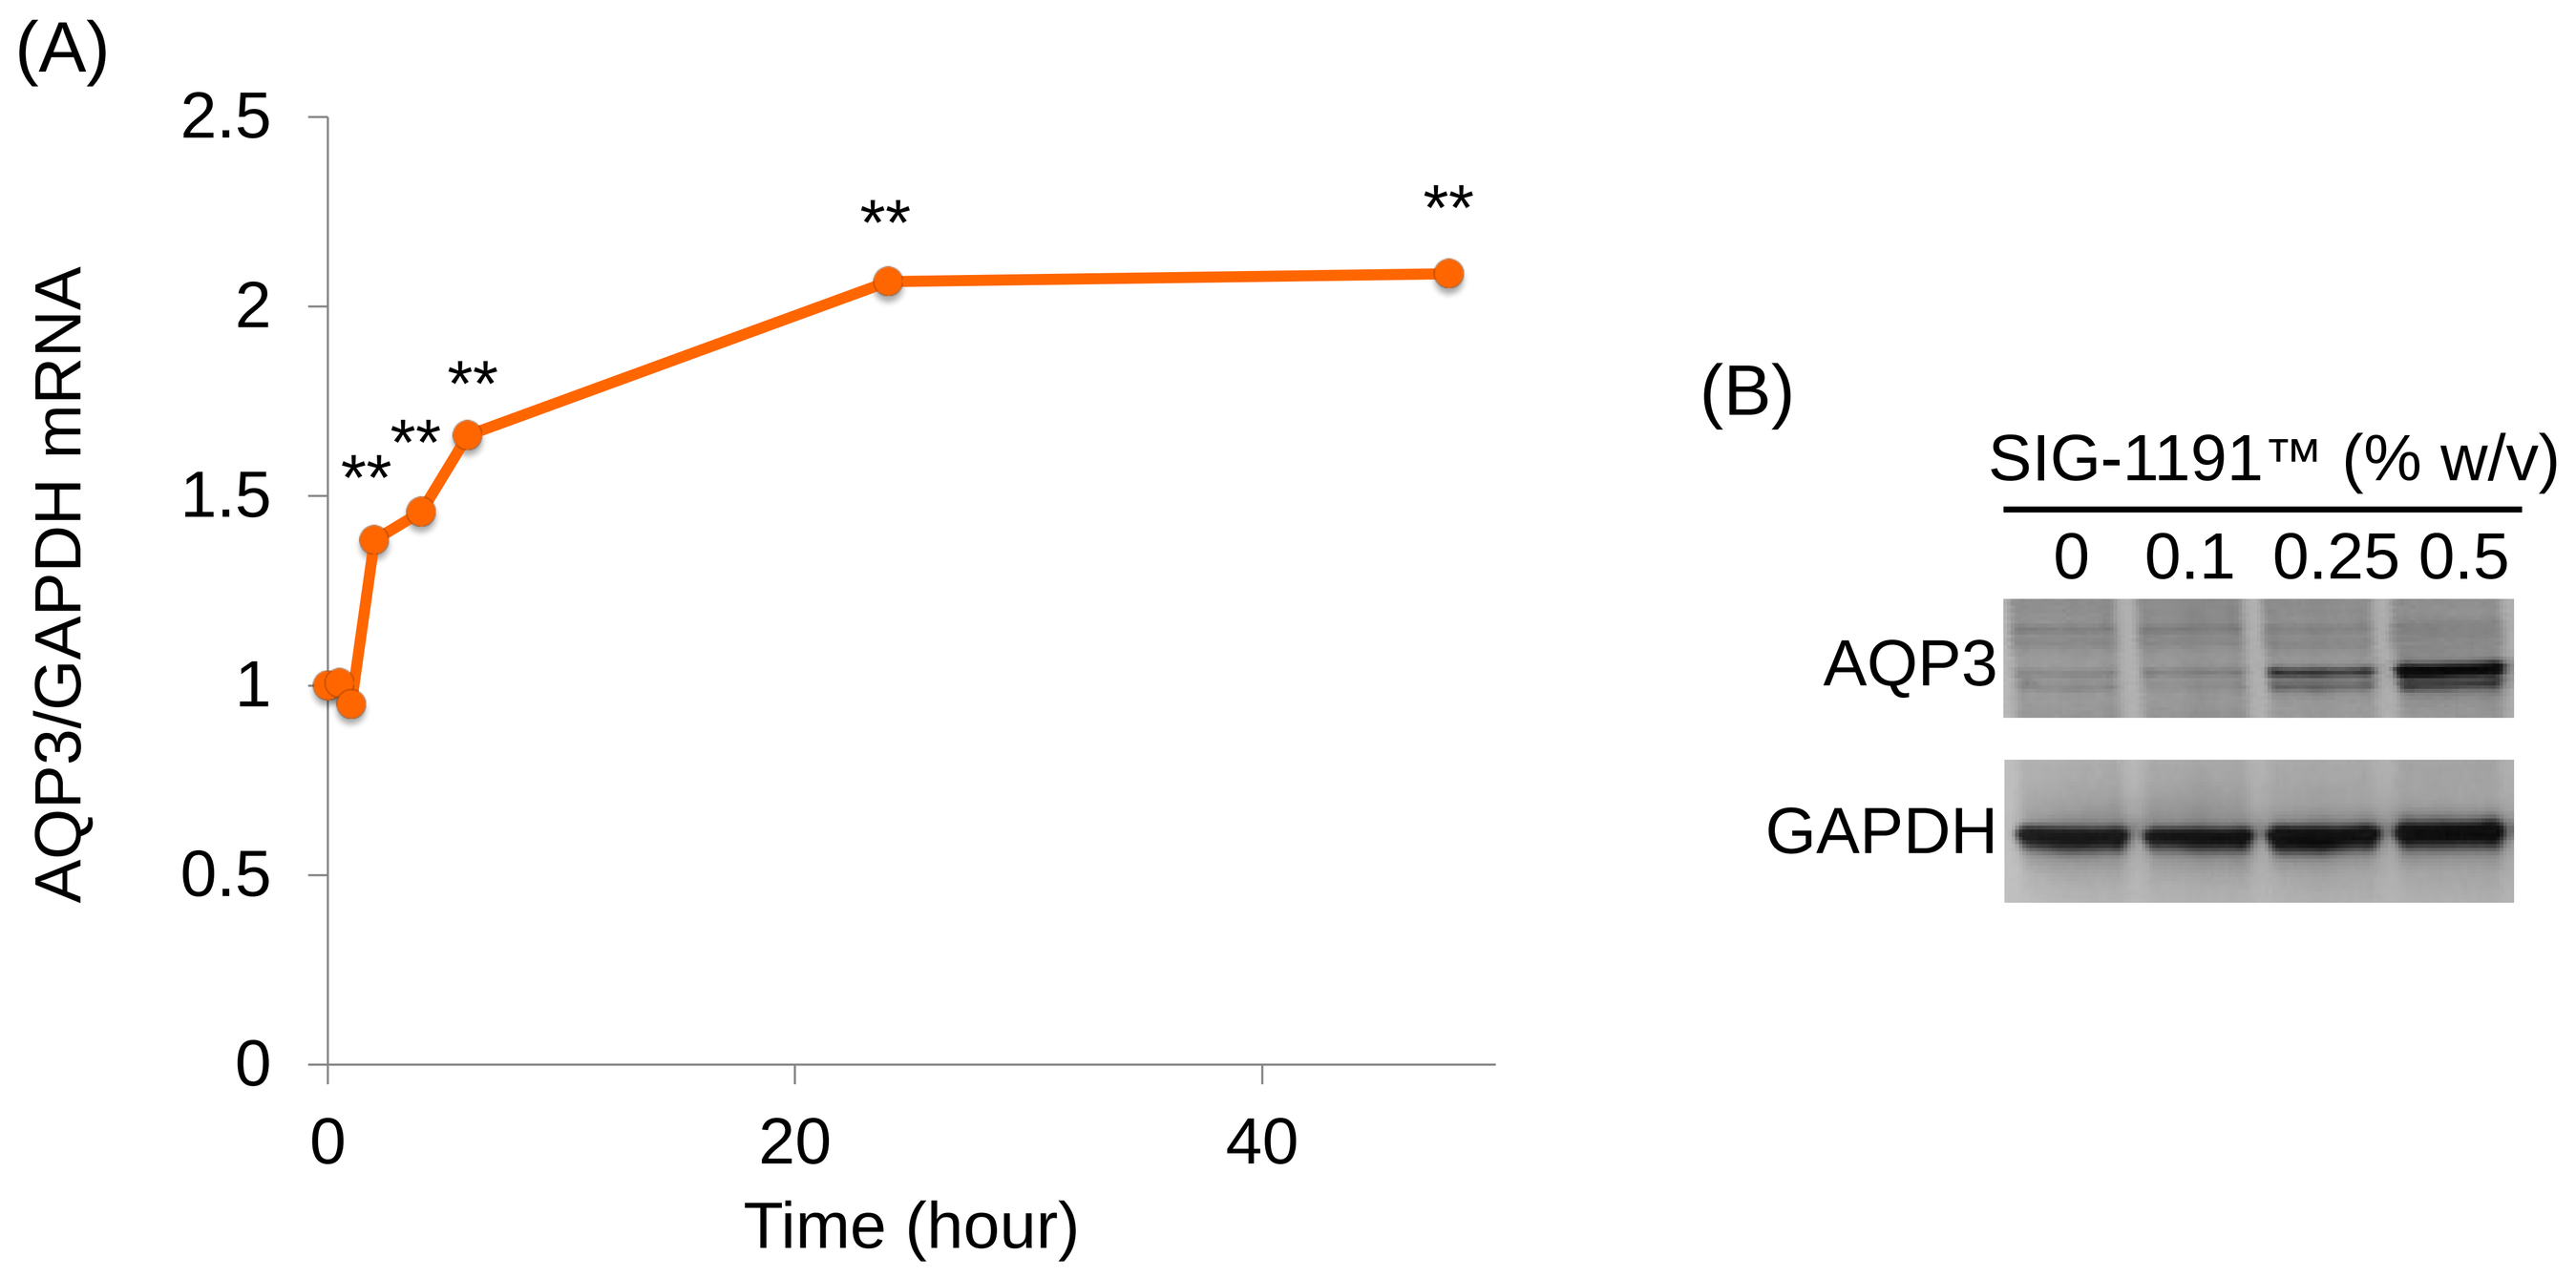
(A)
AQP3/GAPDH mRNA
Time (hour)
0 20 40
0 0.5 1 1.5 2 2.5
**
(B)
SIG-1191™ (% w/v)
0 0.1 0.25 0.5
AQP3
GAPDH

(A) Human Epidermal Keratinocytes were cultured for 24 hours with 10µM SIG-1191™. Total RNA was extracted and qPCR performed for human aquaporin-3 (AQP3) gene expression using GAPDH gene as internal control. (B) EpiDerm-FT™ 3D skin cultures were topically exposed to SIG-1191™ for 7 days and harvested for western blot analysis.

Fig 3. SIG-1191™ stimulates antioxidant activity in dermal fibroblasts

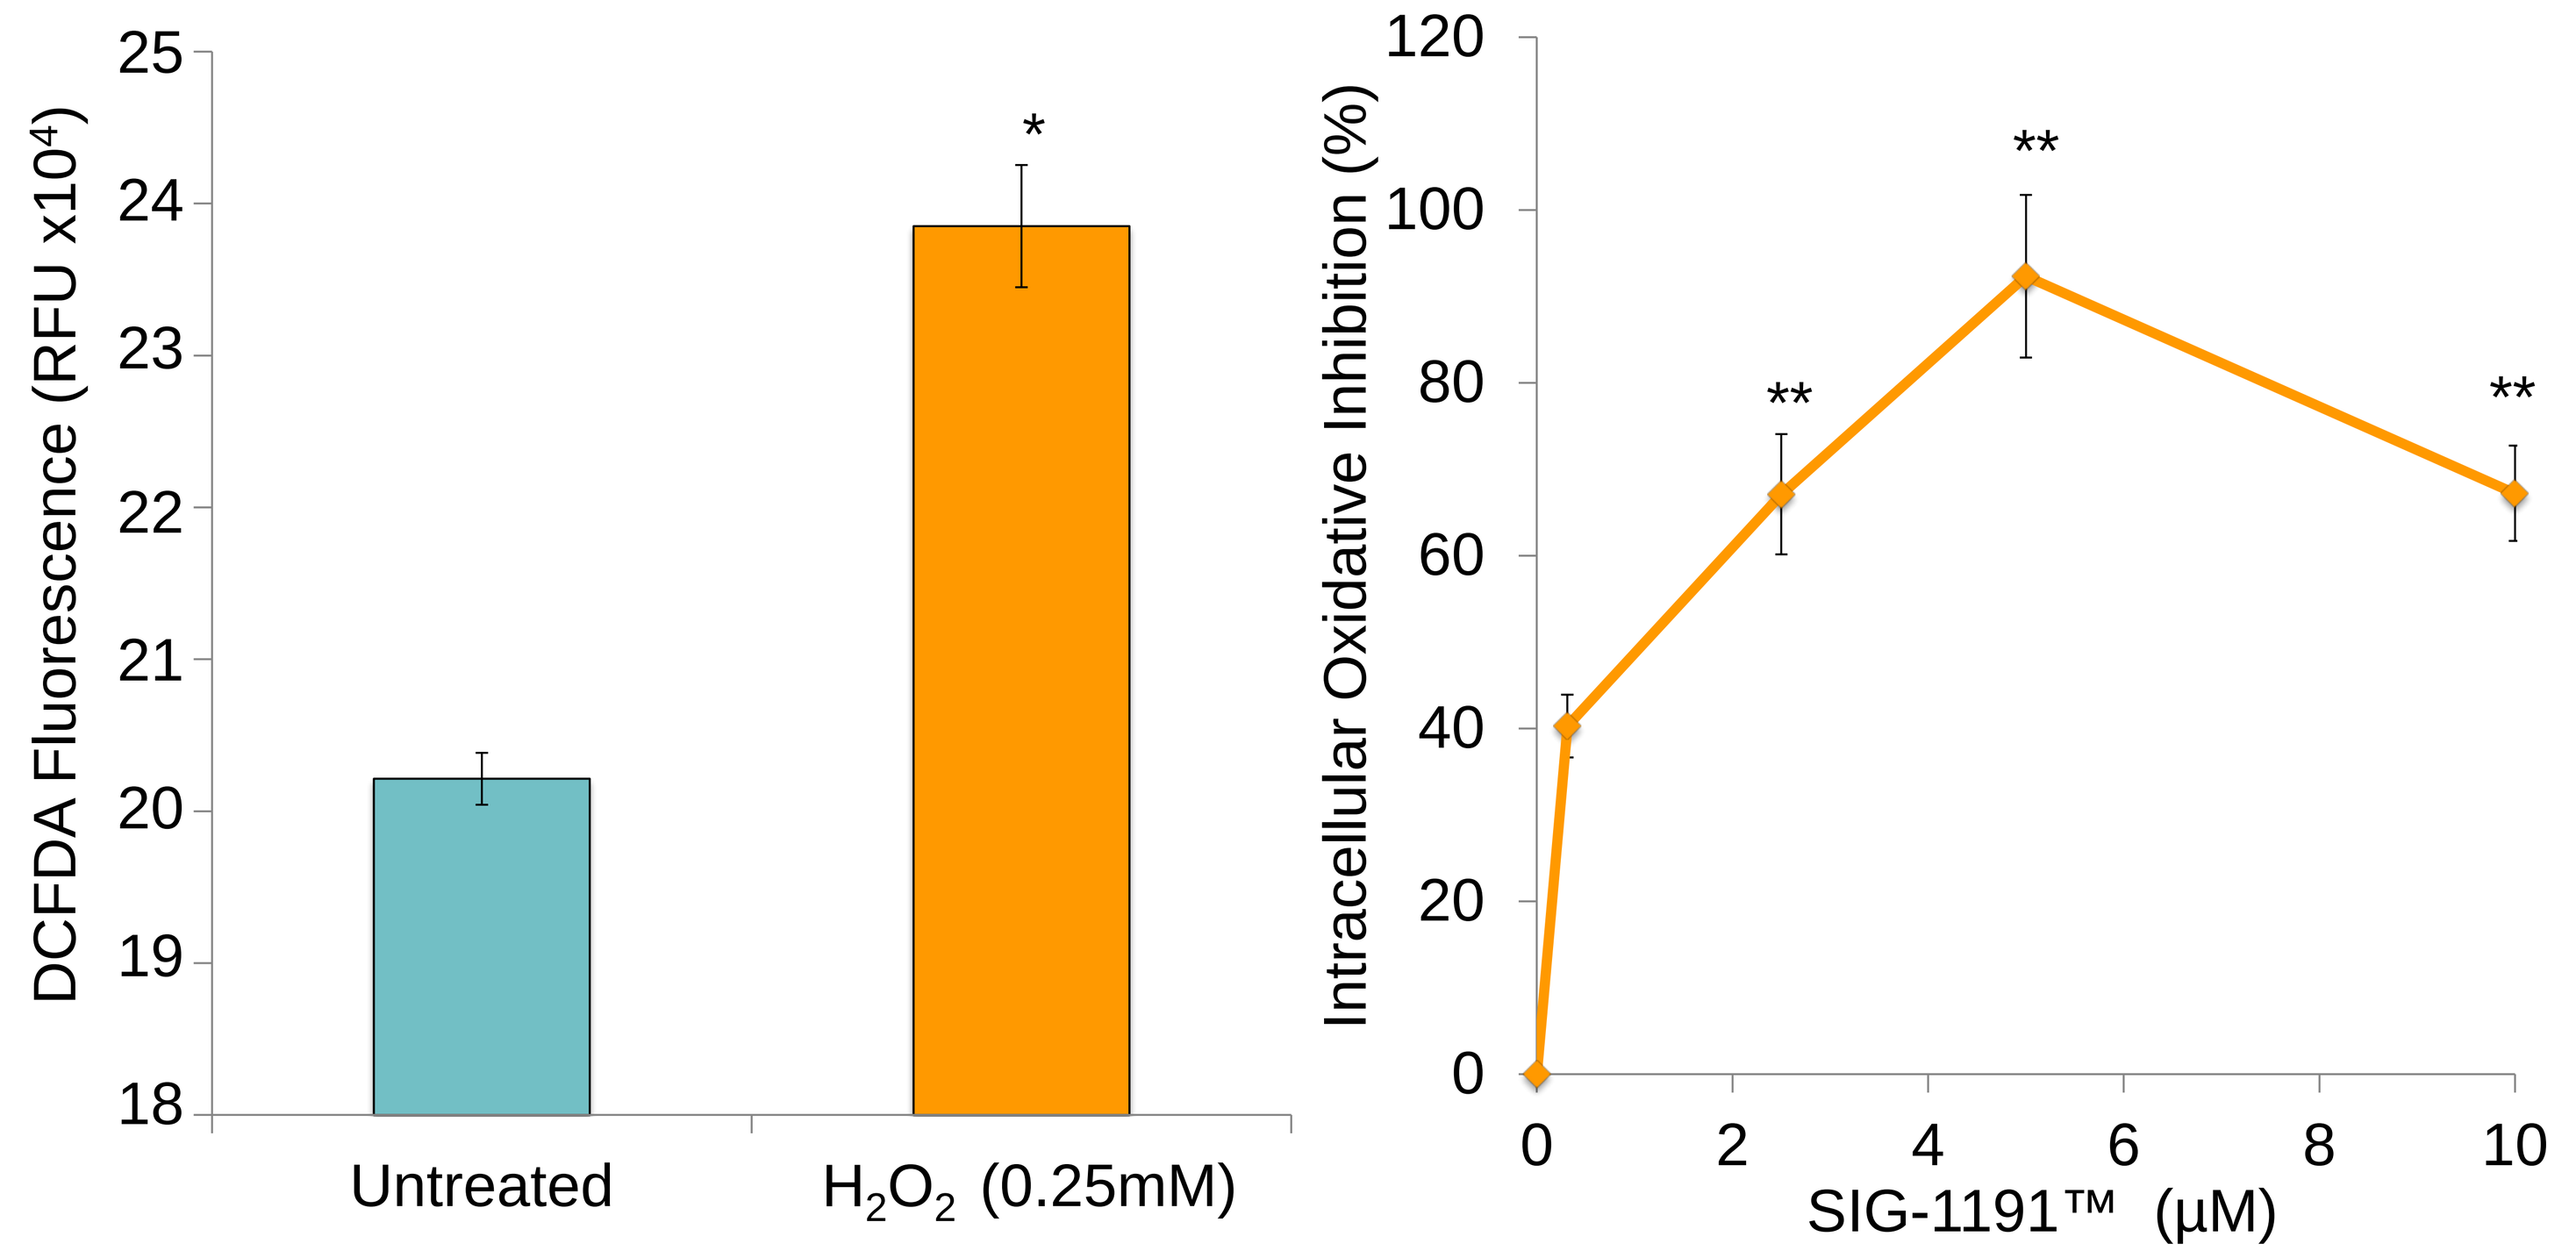
DCFDA Fluorescence (RFU x10⁴)
18 19 20 21 22 23 24 25
Untreated
H_2O_2 (0.25mM)
*
Intracellular Oxidative Inhibition (%)
0 20 40 60 80 100 120
0 2 4 6 8 10
SIG-1191™ (µM)
**

Primary Human Dermal Fibroblasts (HDFs) were incubated with SIG-1191™ (≤ 10µM) for 3 hours. Control cells received vehicle-only. Intracellular oxidative activity was determined using DCFH-DA marker and hydrogen peroxide (0.25 mM) as inducer of oxidative stress.
* p value ≤ 0.05; ** p value ≤ 0.01 by Student *t* test compared to H_2O_2-only treated cells.

Fig 4. SIG-1191™ inhibits UVB-induced cytokines in keratinocytes

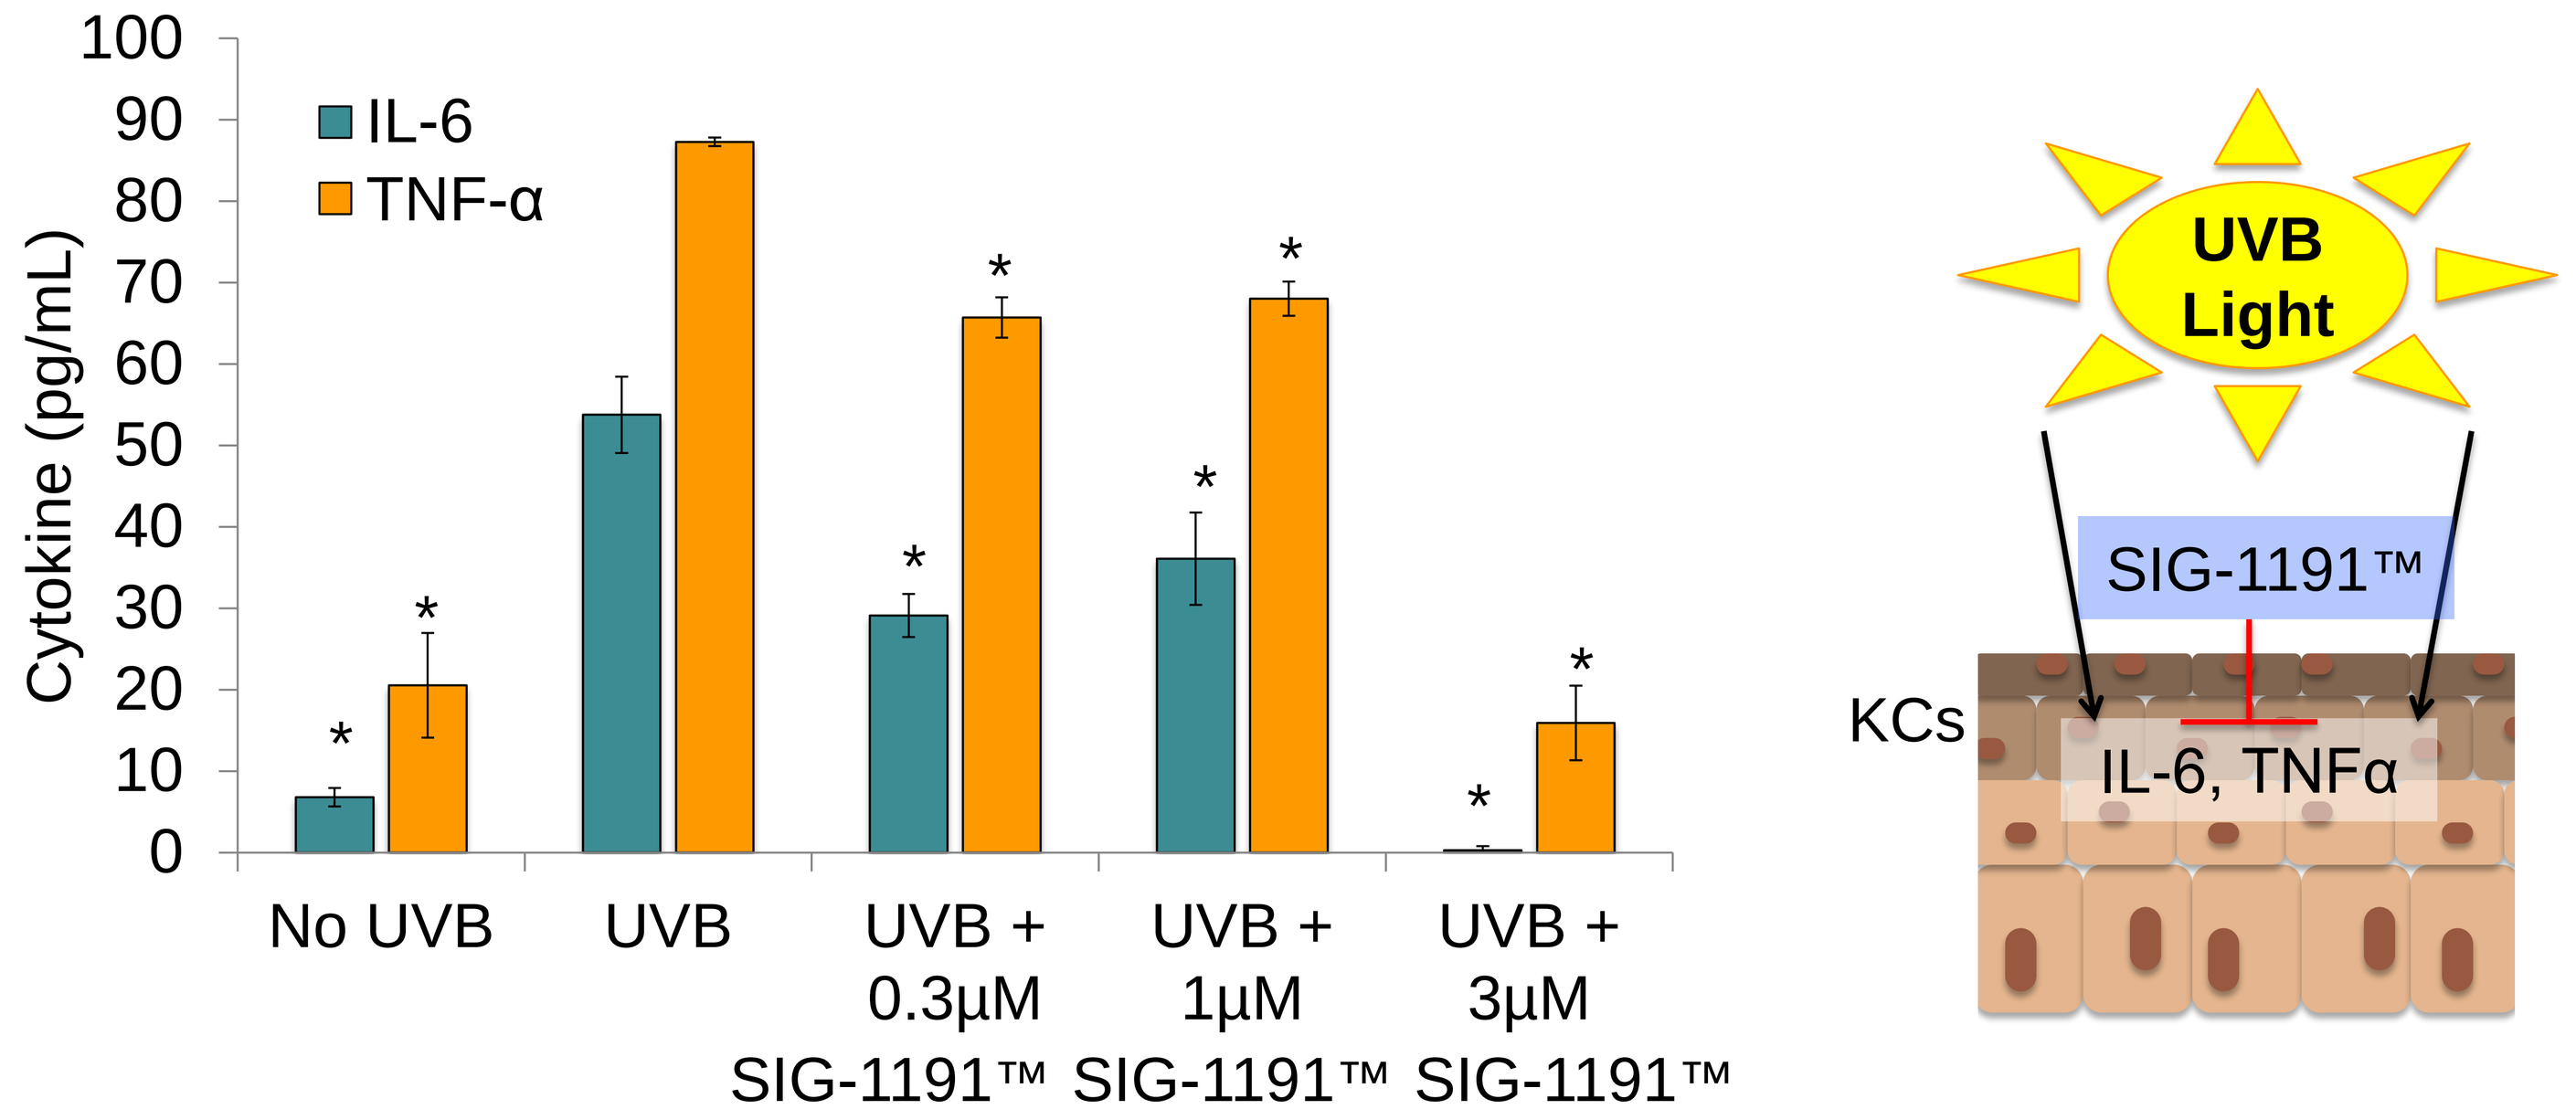
Cytokine (pg/mL)
0 10 20 30 40 50 60 70 80 90 100
IL-6
TNF-α
No UVB
UVB
UVB + 0.3µM SIG-1191™
UVB + 1µM SIG-1191™
UVB + 3µM SIG-1191™
*
UVB Light
SIG-1191™
KCs
IL-6, TNFα

Primary Human Epidermal Keratinocytes (KCs) were seeded in 24-well plates and cultured for 24 hours at 37ºC and 5% CO_2 before treatments. Cells were cultured with SIG-1191™ (0.3-3µM) for 6 hours. The medium was removed, replaced with PBS and cells were irradiated with 25 mJ/cm² UVB. Medium was replaced and supernatants were collected after 24 hours and analyzed by ELISA for IL-6 and TNFα protein levels. *p<0.05 by ANOVA test compared with UVB irradiated cells.

Fig 5. SIG-1191™ increases HO-1 expression in microvascular endothelial cells

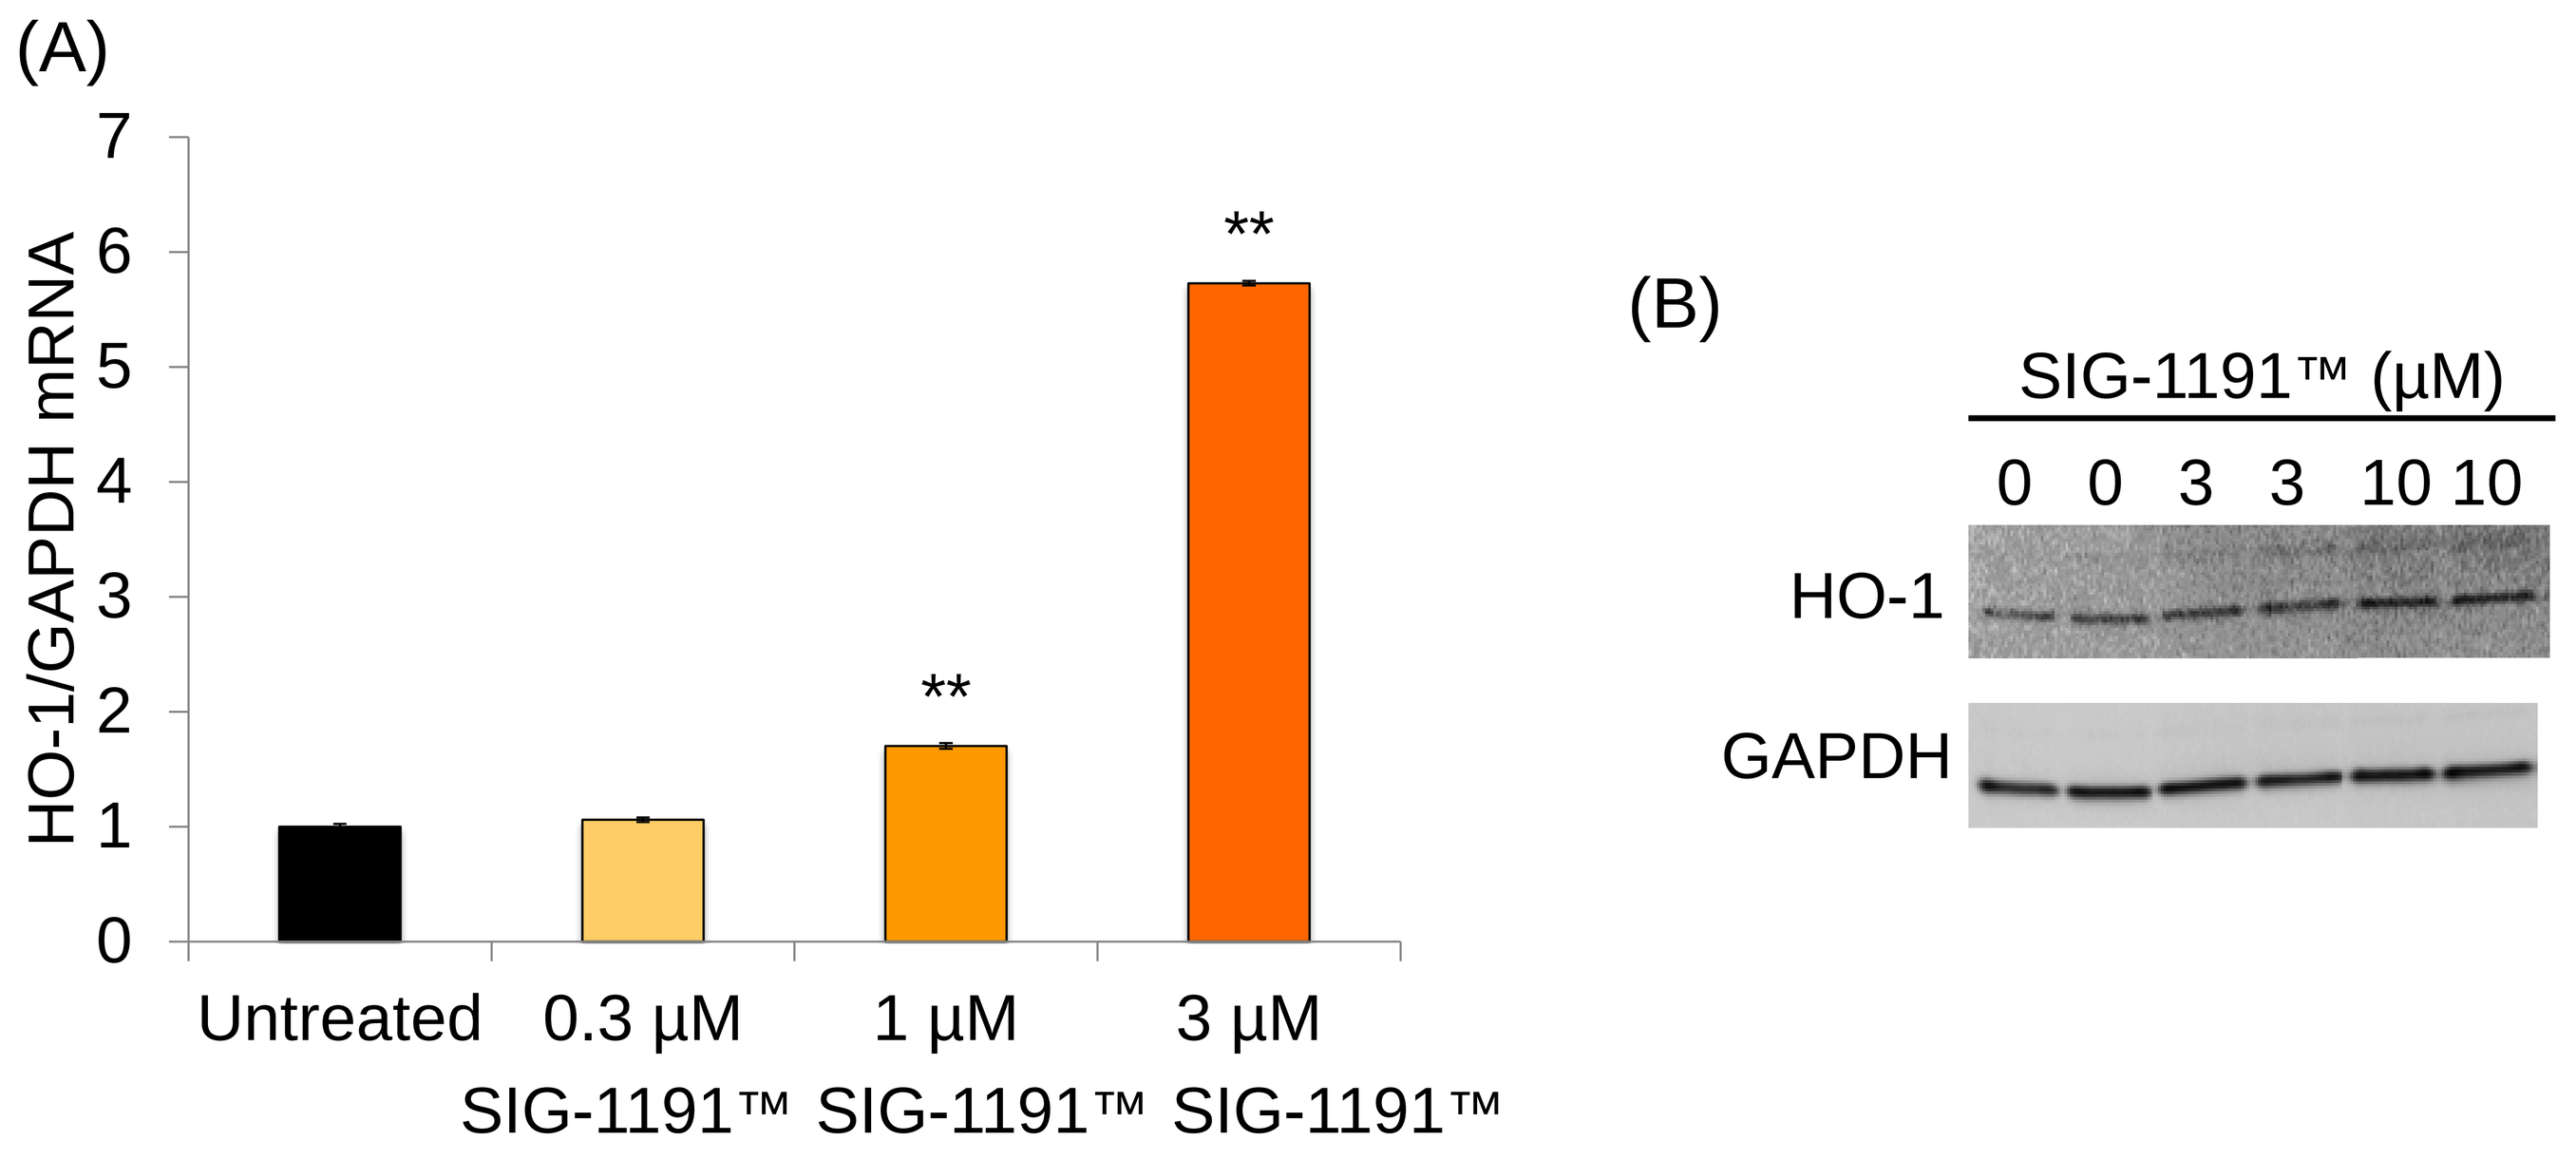
(A)
HO-1/GAPDH mRNA
0 1 2 3 4 5 6 7
Untreated
0.3 µM SIG-1191™
1 µM SIG-1191™
3 µM SIG-1191™
**
(B)
SIG-1191™ (µM)
0 0 3 3 10 10
HO-1
GAPDH

Human Dermal Microvascular Endothelial Cells (HDMECs) were seeded in 6-well plates and pre-cultured for 24 hours at 37ºC and 5% CO_2 before treatments. SIG-1191™ (0.3-3µM) was added to culture media and incubated for an additional 24 hours. Total RNA and protein was extracted for qPCR (A) or western blot analysis (B) for human Heme Oxygenase-1 (HO-1) using GAPDH gene or protein as internal controls. *p<0.05; **p≤0.01 by ANOVA test compared with untreated cells as control.

Fig 6. SIG-1191™ increases HO-1 expression in 3D reconstituted human skin cultures

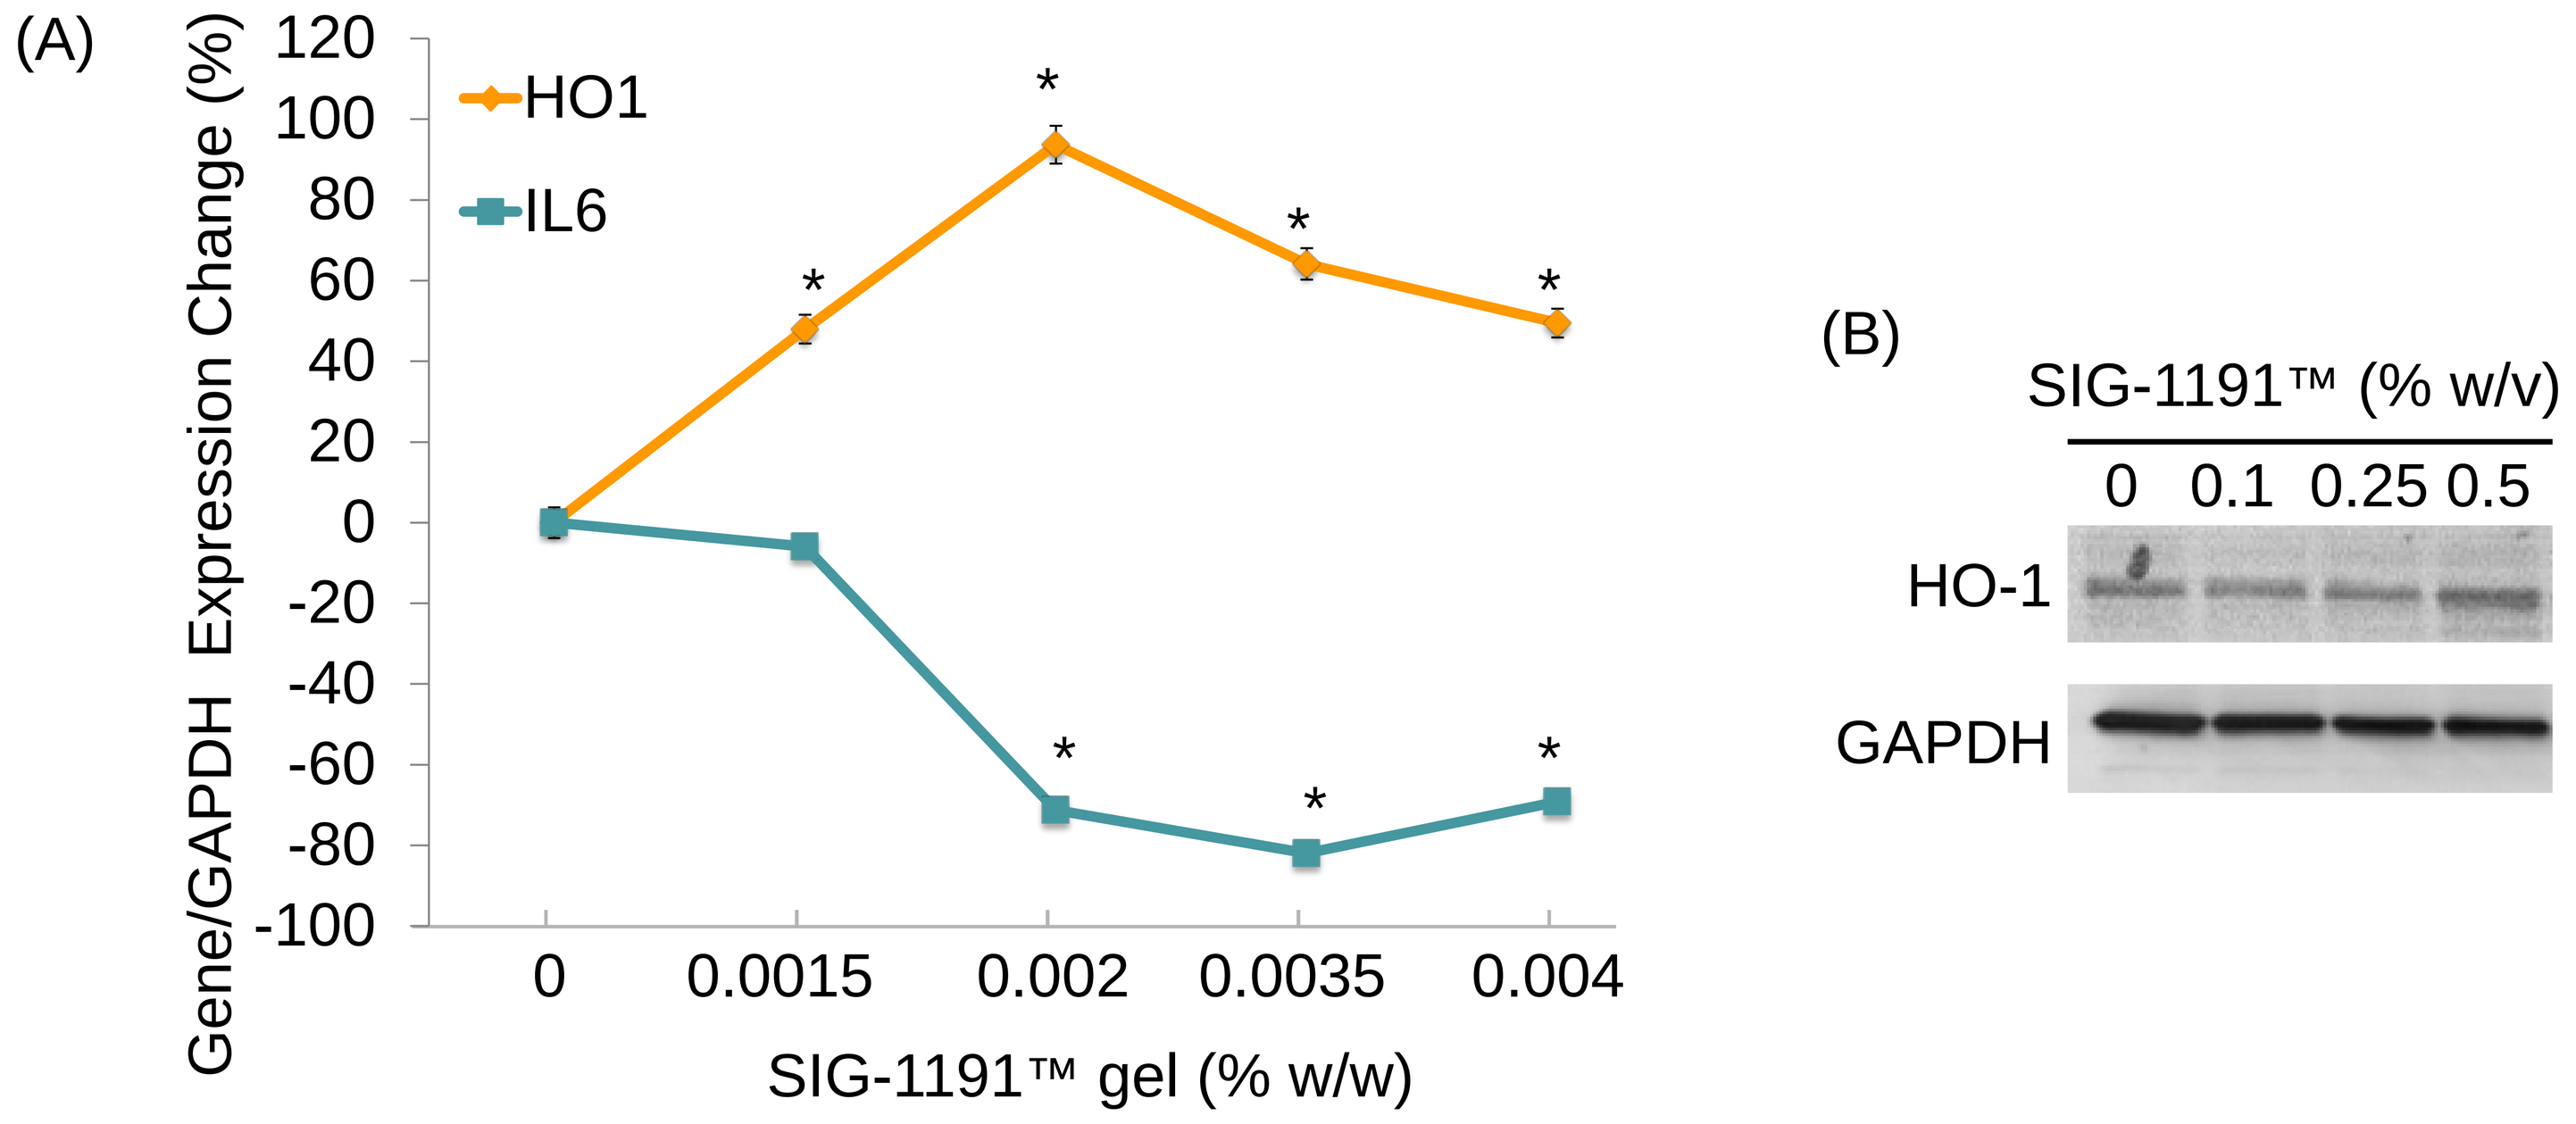
(A)
Gene/GAPDH Expression Change (%)
-100 -80 -60 -40 -20 0 20 40 60 80 100 120
HO1
IL6
0 0.0015 0.002 0.0035 0.004
SIG-1191™ gel (% w/w)
*
(B)
SIG-1191™ (% w/v)
0 0.1 0.25 0.5
HO-1
GAPDH

(A) Reconstituted Human Skin cultured at the air-liquid interface (EpiDerm-FT™; MatTek Corp.) were topically treated with SIG-1191™ formulated in a gel for 24 hours and total RNA was extracted for qPCR analysis of HO-1 and IL-6 gene expression using GAPDH gene as internal control. (B) Cultures were treated with SIG-1191™ formulated in water and analyzed for HO-1 protein expression using GAPDH protein as internal control. *p<0.05 by ANOVA test compared with untreated cells as control.

Fig 7. IHC demonstrates SIG-1191™ promotes HO-1 production in 3D human skin model

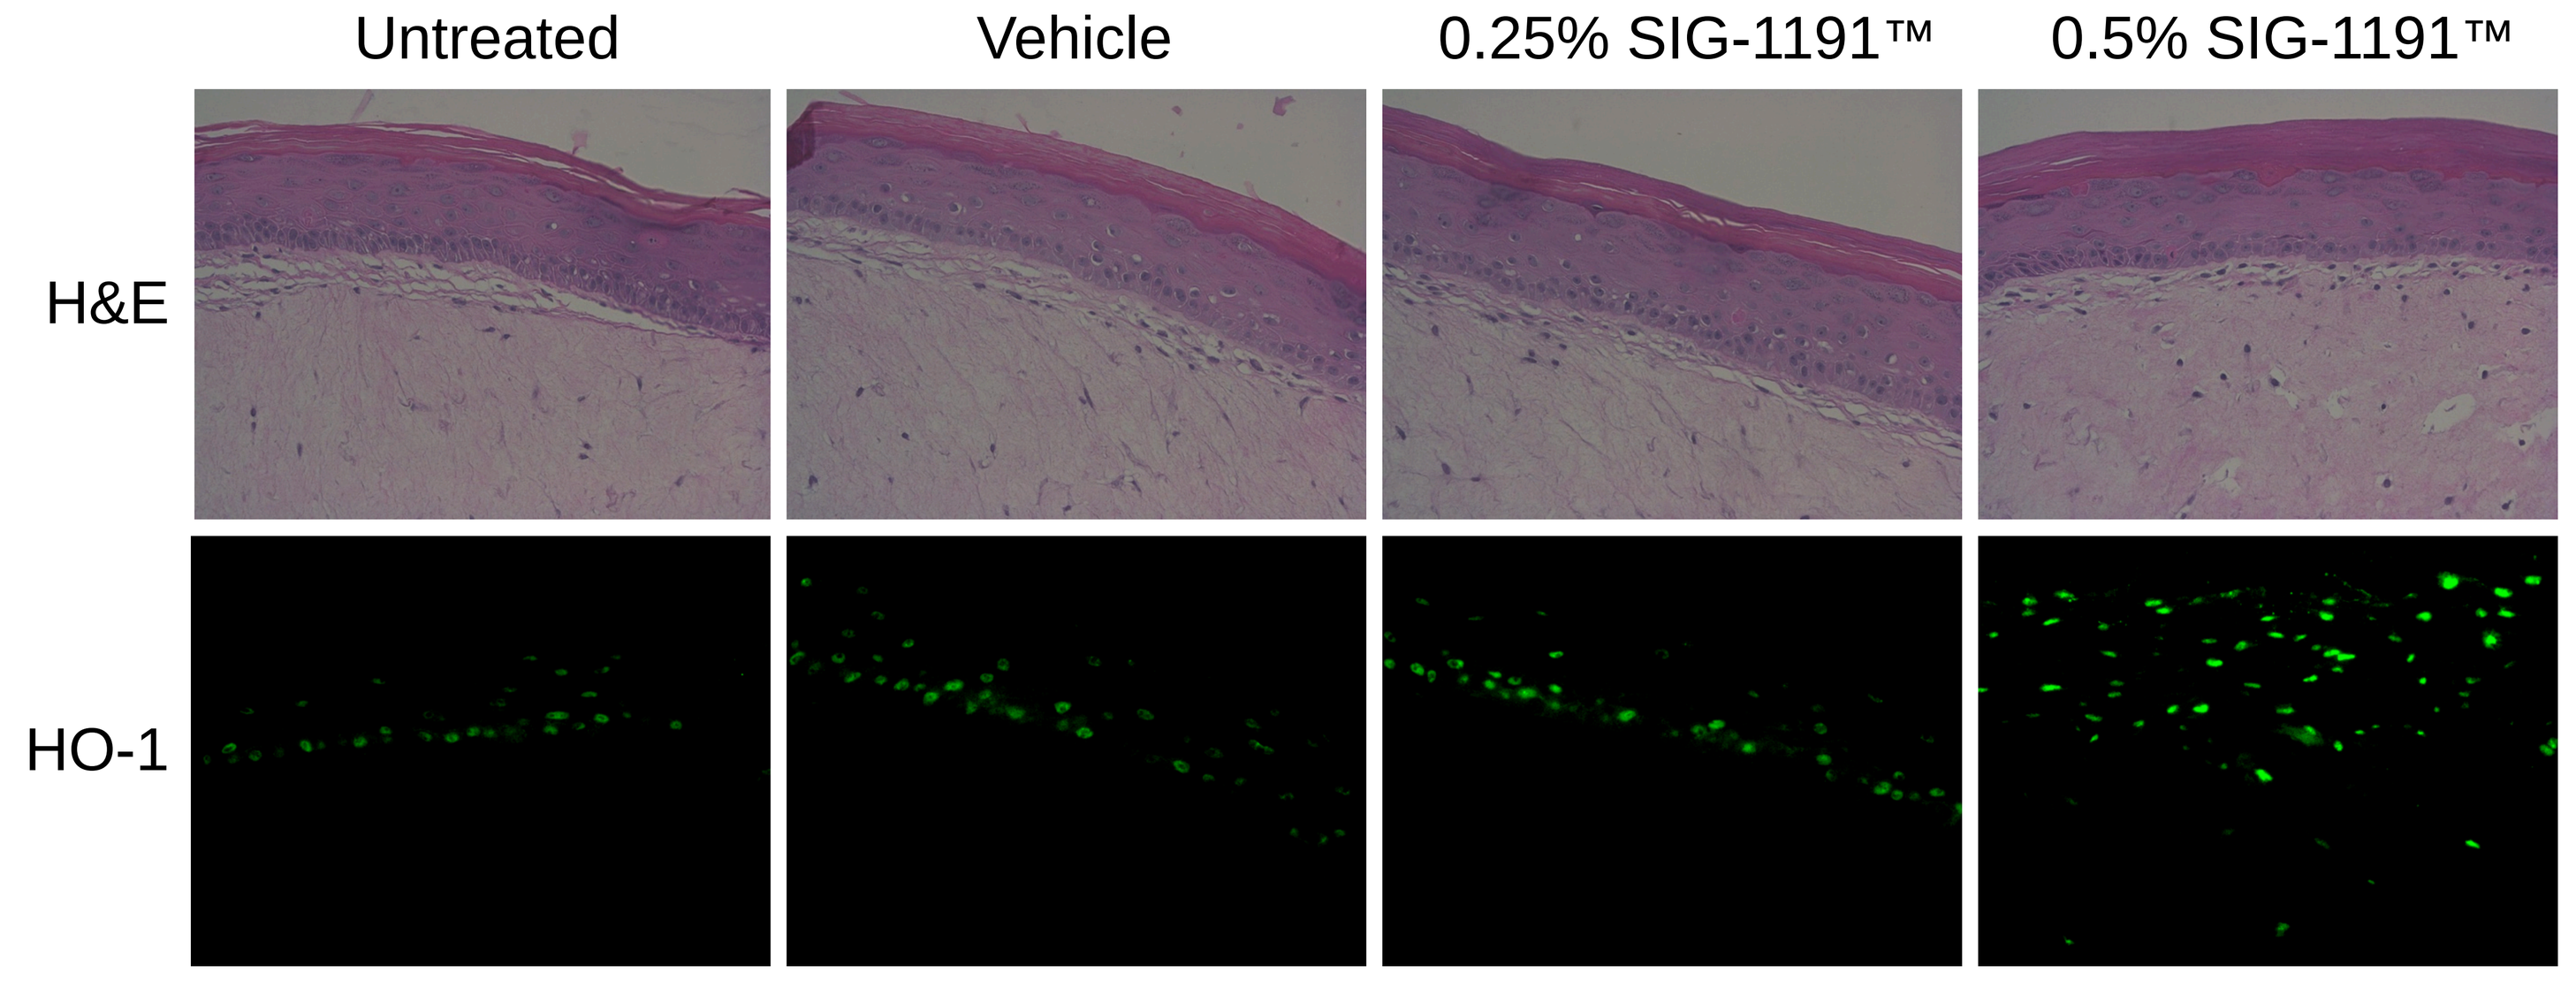
Untreated
Vehicle
0.25% SIG-1191™
0.5% SIG-1191™
H&E
HO-1

EpiDerm-FT™ air-liquid interface cultures were topically treated with 0.25-0.5 % (w/v) of SIG-1191™ for 24 hours. Tissues were subjected to Haematoxylin and Eosin (H&E) staining and immunohistochemistry was performed with anti-HO-1, anti-mouse AlexaFluor®-488 antibodies. HO-1 antibody staining localized to basal epidermal layer and partially to the lowest superbasal layer, predominantly restricted to the cell nucleus in untreated and vehicle exposed cultures. Treatment with SIG-1191™ increased in an apparent dose dependent manner the intensity and distribution of HO-1 staining. Original magnification: x 400.

Summary/Conclusions

- ✧ SIG-1191™ potentially targets skin hydration and aging by modulating Aquaporin-3 (AQP3) expression in both monolayer keratinocytes and 3D skin equivalent cultures.
- ✧ SIG-1191™ demonstrates antioxidant and anti-inflammatory properties *in vitro* reducing intracellular oxidative radicals in human dermal fibroblasts and UVB-induced pro-inflammatory cytokine production in epidermal keratinocytes.
- ✧ Results suggest SIG-1191™ mitigates skin hyperpigmentation affected by heme pigment by increasing Heme oxygenase-1 (HO-1) gene and protein expression as shown in both monolayer dermal microvascular endothelial cells and 3D skin equivalent cultures.
- ✧ In conclusion, these results demonstrate that SIG-1191™ is a novel cosmetic ingredient that potentially promotes skin hydration and reduces suborbital hyperpigmention by increasing Aquaporin-3 and Heme oxygenase-1 levels and through its anti-inflammatory properties.